P2061

Safety outcomes with rezafungin and caspofungin in the treatment of candidaemia and/or invasive candidiasis: Phase 3 data from the ReSTORE trial

A Soriano[1], GR Thompson III[2], OA Cornely[3], B J Kullberg[4], M Kollef[5], J Vazquez[6], PM Honore[7], M Bassetti[8], J Pullman[9], C Dignani[10], AF Das[11], T Sandison[11], PG Pappas[12] on behalf of the ReSTORE trial investigators

[1]Hospital Clínic de Barcelona, IDIBAPS, University of Barcelona, Spain; [2]University of California Davis Medical Center, Davis, CA, USA;[3]University of Cologne, Faculty of Medicine and University Hospital Cologne, Department of Internal Medicine, Excellence Center for Medical Mycology (ECMM), Cologne, Germany; University of Cologne, Faculty of Medicine and University Hospital Cologne, Chair Translational Research, Cologne Excellence Cluster on Cellular Stress Responses in Aging-Associated Diseases (CECAD), Cologne, Germany; University of Cologne, Faculty of Medicine and University Hospital Cologne, Clinical Trials Centre Cologne (ZKS Köln), Cologne, Germany; German Centre for Infection Research (DZIF), Partner Site Bonn-Cologne, Cologne, Germany; [4]Radboud University Medical Center, Nijmegen, The Netherlands; [5]Washington University, St. Louis, MO, USA; [6]Augusta University, Augusta, GA, USA; [7]Brugman University Hospital, Brussels, Belgium; 8University of Genoa, Genoa, Italy; [9]Mercury Street Medical, Butte, MT, USA; [10]PSI-CRO, Durham, NC, USA; [11]Cidara Therapeutics, Inc., San Diego, CA, USA; [12]University of Alabama at Birmingham, Birmingham, AL, USA

INTRODUCTION AND OBJECTIVES

- Candidemia and invasive candidiasis infections are a major cause of morbidity and mortality in hospitals.[1,2] The novel once-weekly echinocandin antifungal, rezafungin for intravenous (IV) injection, offers prolonged half-life (approximately 133 hours) and high front-loaded plasma exposures.[3–5] Rezafungin is in development for the treatment of *Candida* infections and prevention of *Candida*, *Aspergillus*, and *Pneumocystis* infections in allogeneic blood and marrow transplantation.[3–7]
- The current analysis reports safety data from the ReSTORE trial (NCT03667690), examining treatment outcomes with the novel once-weekly echinocandin, rezafungin, and daily caspofungin treatment in people with candidaemia/invasive candidiasis.[7]

METHODS

- ReSTORE comprised a global, randomised, double-blind, double-dummy, Phase 3 non-inferiority trial.
- Adults with candidaemia/invasive candidiasis, diagnosed by systemic manifestations and mycological confirmation, were randomised to receive once-weekly IV rezafungin (Week 1: 400 mg; Weeks 2–4: 200 mg) or once daily caspofungin (Day 1: 70 mg; Days 2–28: 50 mg) for ≥14 days (≤4 weeks).
- Vital signs and laboratory data were collected throughout the study. Safety outcomes included reporting of treatment-emergent adverse events (TEAEs) and serious adverse events (SAEs). The analysis examined the Safety Population (all participants who received ≥1 dose of study drug).

RESULTS

Changes in vital signs and laboratory values during the study period (Safety Population)

- The Safety Population included 98 patients in each treatment arm. Table 1 shows clinically significant changes in key vital sign and laboratory parameters during the study period for the Safety Population.
- Overall, 29.2% (rezafungin arm) and 26.3% (caspofungin arm) demonstrated reductions in systolic blood pressure ≥20 mmHg to a value of ≤90 mmHg. Heart rate was raised by ≥15 bpm to a value ≥120 bpm for 24.0% (rezafungin arm) and 26.3% (caspofungin arm). QTcF values increased by ≥30 msec in 12.0% (rezafungin arm) and 17.5% (caspofungin arm).
- Increases in aspartate aminotransferase and/or alanine aminotransferase, total bilirubin and alkaline phosphatase were similar between the treatment groups. Nephrotoxicity occurred in 9.3% of rezafungin-treated subjects and 18.8% of caspofungin-treated subjects during the study period.

Table 1. Clinically significant change in key vital signs and laboratory parameters from baseline (ReSTORE trial Safety Population)

Reported change in key vital sign or laboratory value, n/N1 (%)	Rezafungin (400/200 mg) (N=98)	Caspofungin (70/50 mg) (N=98)
Systolic blood pressure (mmHg)		
Increase of ≥20 mmHg (≥180 mmHg)	12/96 (12.5)	16/95 (16.8)
Decrease of ≥20 mmHg (≤90 mmHg)	28/96 (29.2)	25/95 (26.3)
Diastolic blood pressure (mmHg)		
Increase of ≥15 mmHg (≥105 mmHg)	11/96 (11.5)	12/95 (12.6)
Decrease of ≥15 mmHg (≤50 mmHg)	27/96 (28.1)	21/95 (22.1)
Heart rate (bpm)		
Increase of ≥15 bpm (≥120 mmHg)	23/96 (24.0)	25/95 (26.3)
Decrease of ≥15 bpm (≤50 mmHg)	8/96 (8.3)	12/95 (12.6)
Temperature (°C)		
Increase of ≥1°C (>38°C)	20/96 (20.8)	14/95 (14.7)
Decrease of ≥1°C (<36 °C)	29/96 (30.2)	32/95 (33.7)
Respiratory rate (breaths/minute)		
Increase of ≥10 breaths/minute (≥30 breaths/minute)	17/92 (18.5)	12/88 (13.6)
Decrease of ≥4 breaths/minute (≤8 breaths/minute)	4/92 (4.3)	3/88 (3.4)
Aggregate QTcF interval (msec)		
Increase of ≥30 msec	9/75 (12.0)	14/80 (17.5)
Increase of ≥60 msec	3/75 (4.0)	8/80 (10.0)
Alanine aminotransferase (ULN)		
Increase of >3 x ULN	10/ 97 (10.3)	14/ 96 (14.6)
Aspartate aminotransferase (ULN)		
Increase of >3 x ULN	16/ 97 (16.5)	17/ 96 (17.7)
Total bilirubin (ULN)		
Increase of >1.5 x ULN	20/ 97 (20.6)	21/ 96 (21.9)
Alkaline phosphatase (ULN)		
Increase of >1 x ULN	32/ 96 (33.3)	27/ 95 (28.4)

The Safety Population included all subjects who had received ≥1 dose of study drug.
Abbreviations: bpm, beats per minute; n, number of subjects demonstrating the relevant change in vital sign, N1, number of subjects with a baseline and at least one post-baseline measurement; ULN, upper limit of normal.

Treatment-related adverse events

- Sixteen subjects reported treatment-related TEAEs with rezafungin (including 5 related to IV saline/placebo administration) and 9 with caspofungin (Table 2). Adverse events (AEs) of special interest included Grade 1 tremors (2 rezafungin-treated subjects), which were resolved during the study period. Grade 2 peripheral neuropathy (1 subject) and polyneuropathy (1 subject) were reported in the caspofungin group. Both remained unresolved during the study period.
- Treatment-related SAEs included reactions related to IV placebo infusion (rezafungin arm; considered an AE of special interest) and anaphylactic shock (caspofungin arm).
- Early treatment-related TEAEs occurred within the first 4 days (Figure 1). Treatment-related TEAEs remained low during Week 2 of rezafungin and caspofungin treatment. Table 3 shows the treatment-related events reported during the first 7 days.

RESULTS (CONTINUED)

Table 2. ReSTORE trial treatment-related safety data (Safety Population)

	Rezafungin (400/200 mg) (N=98)	Caspofungin (70/50 mg) (N=98)
TEAEs affecting ≥10% of Safety Population (preferred term)		
Pyrexia	14 (14.3)	5 (5.1)
Hypokalaemia	13 (13.3)	9 (9.2)
Pneumonia	10 (10.2)	3 (3.1)
Septic shock	10 (10.2)	9 (9.2)
Subjects with ≥1 drug-related TEAE, n (%)*	16 (16.3)	9 (9.2)
Subjects with ≥1 SAE, n (%)	55 (56.1)	52 (53.1)
Subjects with ≥1 drug-related SAEs (preferred term), n (%)	2 (2.0)	3 (3.1)
Infusion-related reaction†	1 (1.0)	0
Urticaria	1 (1.0)	0
Elevated transaminase levels	0	1 (1.0)
Liver injury	0	1 (1.0)
Anaphylactic shock	0	1 (1.0)
Subjects with AEs of special interest (preferred term), n (%)	6 (6.1)	3 (3.1)
Adverse drug reaction	1 (1.0)	0
Hypersensitivity reaction	1 (1.0)	0
Anaphylactic shock	0	1 (1.0)
Infusion-related reaction†	3 (3.1)	0
Tremor	2 (2.0)	0
Peripheral neuropathy	0	1 (1.0)
Polyneuropathy	0	1 (1.0)

The Safety Population included all subjects who had received ≥1 dose of study drug. *Further investigation revealed that 5 TEAEs believed to be related to rezafungin treatment were associated with the IV saline placebo (for rezafungin) administration. †Two of the four infusion-related reactions observed in the rezafungin group (including the one drug-related SAE) were determined to have occurred in subjects receiving placebo infusions.
Abbreviations: AEs, adverse events; IV, intravenous; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

Figure 1. Incidence of early treatment-related TEAEs (ReSTORE trial Safety Population)

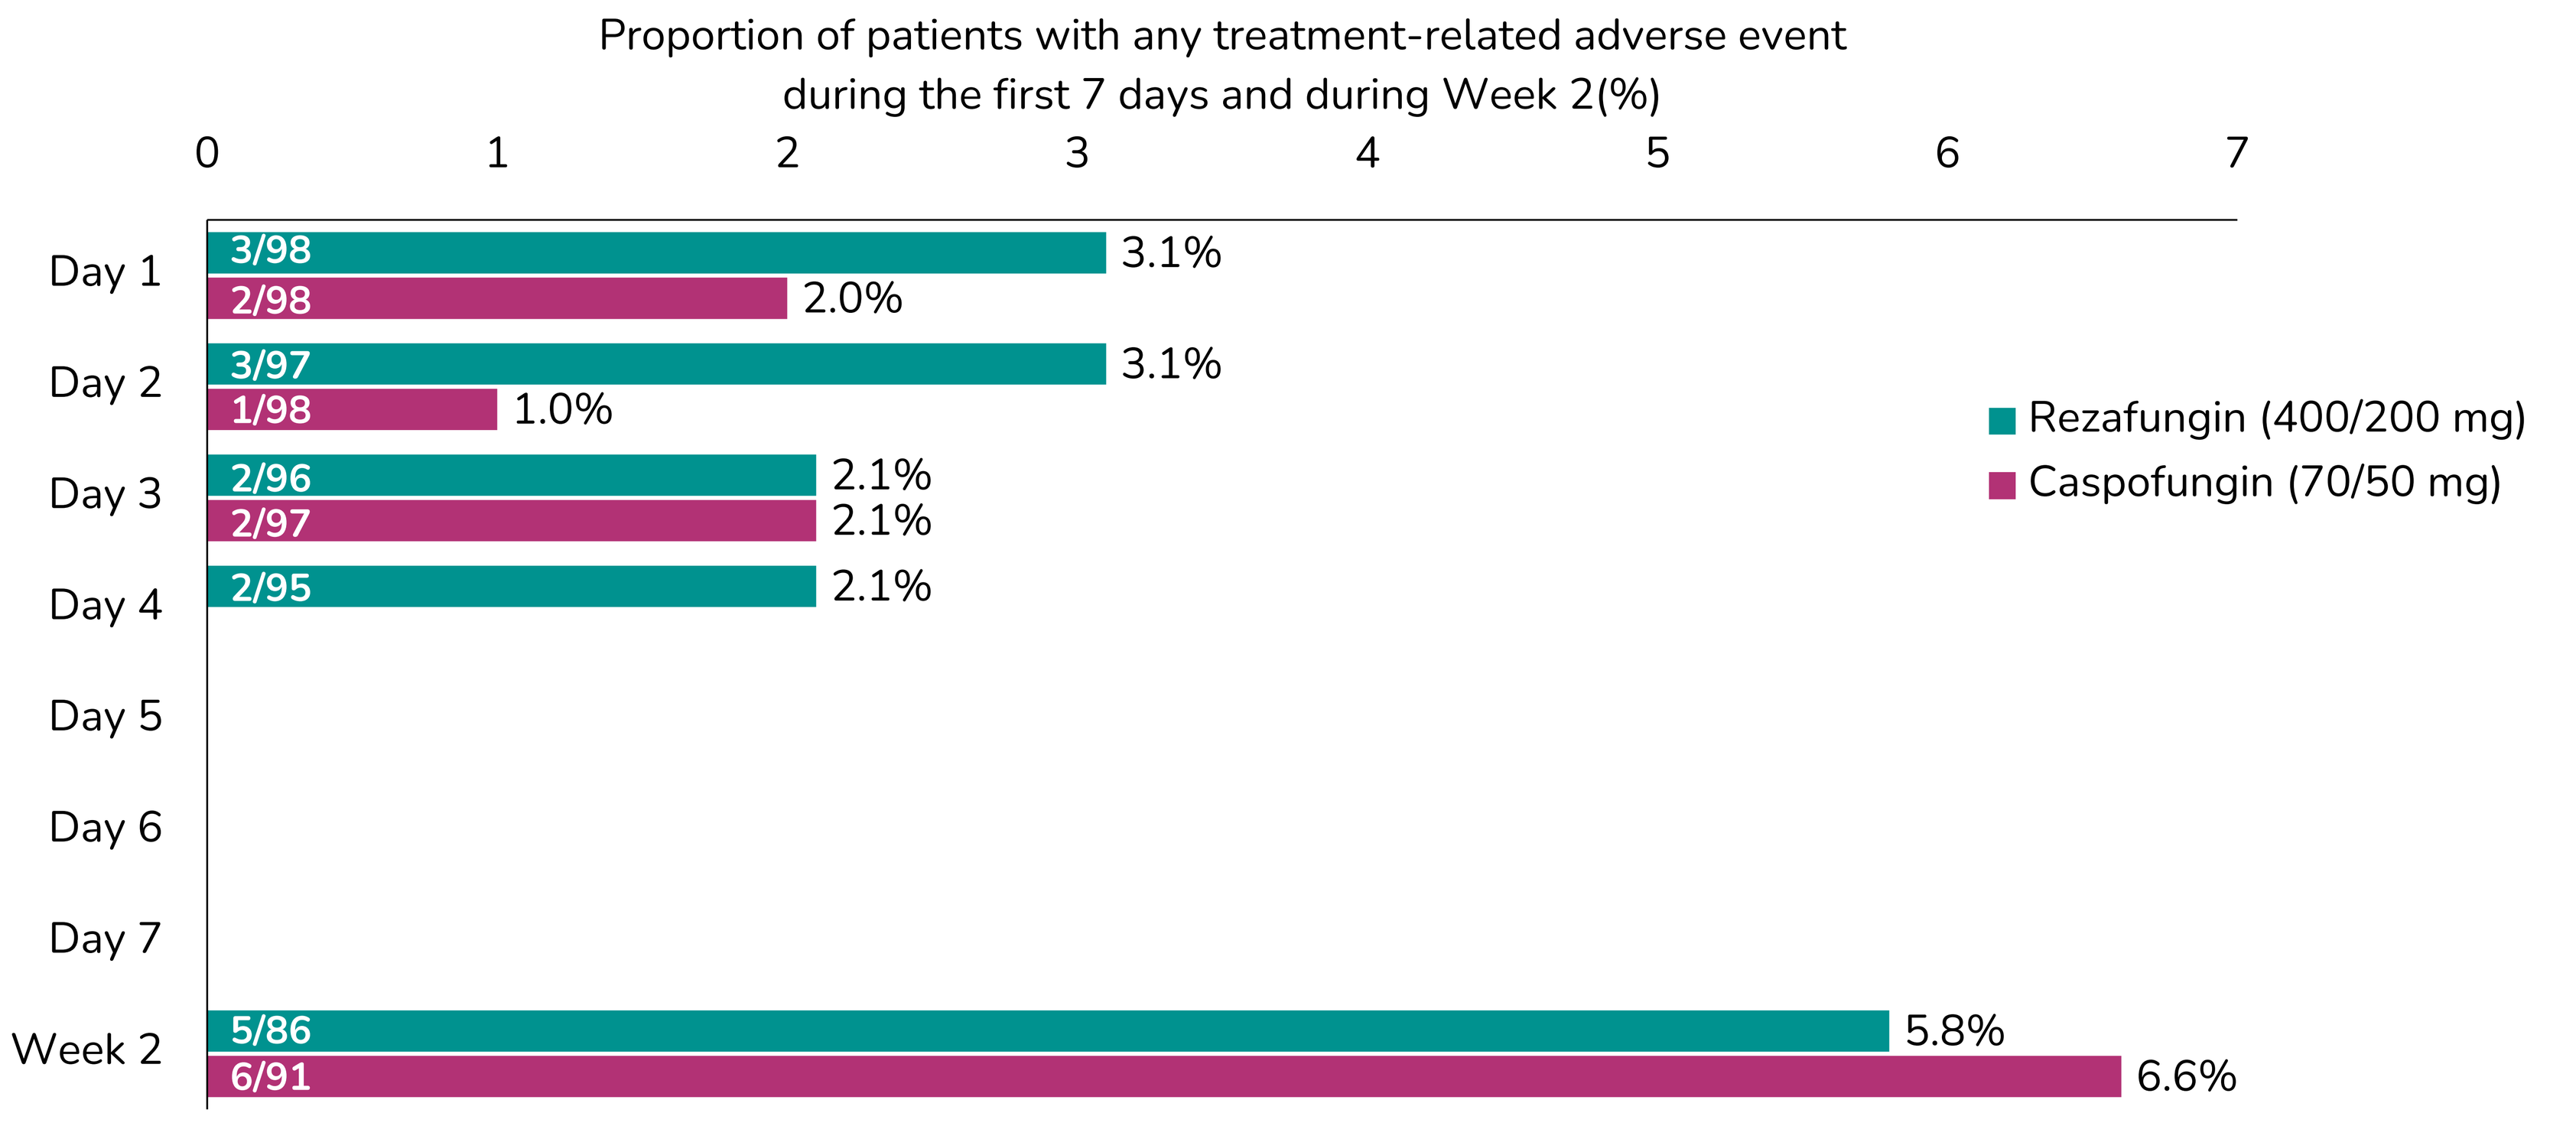
Proportion of patients with any treatment-related adverse event during the first 7 days and during Week 2(%)
0
1
2
3
4
5
6
7
Day 1
3/98
3.1%
2/98
2.0%
Day 2
3/97
3.1%
1/98
1.0%
Day 3
2/96
2.1%
2/97
2.1%
Day 4
2/95
2.1%
Day 5
Day 6
Day 7
Week 2
5/86
5.8%
6/91
6.6%
Rezafungin (400/200 mg)
Caspofungin (70/50 mg)

The Safety Population included all subjects who had received ≥1 dose of study drug.

Table 3. Treatment-related TEAEs occurring during the first 7 days (ReSTORE trial Safety Population)

	Rezafungin (400/200 mg) (N=98)	Caspofungin (70/50 mg) (N=98)
Subjects with ≥1 drug-related TEAE during the first 7 days, n (%)		
Sinus tachycardia	1 (1.0)	0
Vomiting	0	1 (1.0)
Adverse drug reaction	1 (1.0)	0
Hyperbilirubinemia	1 (1.0)	0
Anaphylactic shock	0	1 (1.0)
Pneumonia	1 (1.0)	0
Infusion related reaction†	3 (3.1)	0
Blood alkaline phosphatase increased	1 (1.0)	0
Hepatic enzyme increased	1 (1.0)	0
Hyperphosphatemia	1 (1.0)	0
Hypomagnesaemia	1 (1.0)	0
Decreased appetite	0	1 (1.0)
Coordination abnormal	0	1 (1.0)
Headache	0	1 (1.0)
Nystagmus	0	1 (1.0)
Wheezing	1 (1.0)	0
Erythema	1 (1.0)	0

The Safety Population included all subjects who had received ≥1 dose of study drug. †Two infusion-related reactions in the rezafungin group were determined to have occurred in subjects receiving placebo infusions.

CONCLUSION

- Safety outcomes from the ReSTORE trial indicated that rezafungin had a similar safety profile to caspofungin and the high front-loading dose used for rezafungin did not have any additional impact on safety outcomes.
- Treatment-related TEAEs and SAEs were uncommon in both treatment groups and changes relating to vital signs were similar across groups.

Disclosures

A Soriano: grant from Gilead Sciences; consulting fees and honoraria from Angelini, Gilead, Menarini, MSD, and Shionogi, outside of the submitted work; and grants, consulting fees, honoraria, and support attending meetings from Pfizer, outside of the submitted work. **GR Thompson:** grants and consulting fees from Amplyx, Astellas, Cidara, F2G, and Manye; grants from Merck; and data safety monitoring board membership for Pfizer, outside of the submitted work. **OA Cornely:** reports grants or contracts from Amplyx, Basilea, Bundesministerium für Bildung und Forschung, Cidara, German Center for Infection Research, European Union Directorate-General for Research and Innovation (101037867), F2G, Gilead, Matinas, MedPace, MSD, Mundipharma, Octapharma, Pfizer, and Scynexis; consulting fees from AbbVie, Amplyx, Biocon, Biosys, Cidara, Da Volterra, Gilead, IQVIA, Janssen, Matinas, MedPace, Menarini, Molecular Partners, Noxxon, Octapharma, Pardes, Pfizer, Pharma Support America, Scynexis, and Seres; honoraria from Abbott, AbbVie, Al-Jazeera Pharmaceuticals, Astellas, Gilead, Grupo Biotoscana/ United Medical/Knight, Hikma, MedScape, MedUpdate, Merck/MSD, Mylan, Noscendo, Pfizer, and Shionogi; payment for expert testimony from Cidara; data safety monitoring board or advisory board membership for Actelion, Allecra, Cidara, Entasis, IQVIA, Janssen, MedPace, Paratek, Pharma Support America, Pulmocide, Shionogi, and The Prime Meridian Group; a patent at the German Patent and Trade Mark Office (DE 10 2021 113 007.7); stocks from CoRe Consulting; and is a board member of German Society for Haematology and Medical Oncology, Deutsche Gesellschaft für Information und Wissen, ECMM European Confederation of Medical Mycology, International Society for Human & Animal Mycology, Mycoses Study Group–Education and Research Consortium, and Wiley, outside of the submitted work. **BJ Kullberg:** independent data review committee membership for Cidara. GRT reports grants and consulting fees from Amplyx, Astellas, Cidara, F2G, and Manye; grants from Merck; and data safety monitoring board membership for Pfizer, outside of the submitted work. **M Kollef:** grants from Barnes-Jewish Hospital Foundation and consulting fees from Merck, Pfizer, and Shionogi, outside of the submitted work. **J Vazquez:** consulting fees from and membership of data safety monitoring board or advisory board for F2G and consulting fees from Cidara and Scynexis, outside of the submitted work. **M Bassetti:** honoraria from and membership of data safety monitoring board or advisory board for Angelini, Cidara, Gilead, Menarini, MSD, Pfizer, and Shionogi, outside of the submitted work. **AF Das:** consulting fees from Cidara. **PG Pappas:** grants from and data review committee membership for Cidara; grants from Astellas, Scynexis, and Merck; and advisory board membership for F2G, outside of the submitted work. **T Sandison:** employee of and stocks in Cidara. All other authors declare no competing interests.

33rd ECCMID EUROPEAN CONGRESS OF CLINICAL MICROBIOLOGY AND INFECTIOUS DISEASES

Copenhagen, Denmark
15–18 April 2023

References

1. Pappas PG, et al. Clin. Infect. Dis. 2003;37:634–643; **2.** Kullberg BJ et al. N. Engl. J. Med. 2015;373:1445–1456; **3.** Sandison T, et al. Antimicrob Agents Chemother.2017;61:e01627–16; **4.** Krishnan BR, et al. J Antibiot.2017;70:130–135; **5.** Lakota EA, et al. Antimicrob Agents Chemother.2017;61:e00758-17; **6.** Thompson GR, et al. Clin Infect Dis.2020:ciaa1380; **7.** Thompson GR, et al. Lancet.2022:S0140-6736(22)02324-8.

Funding

ReSTORE trial: co-funded by Cidara Therapeutics and Mundipharma. **STRIVE study:** Cidara Therapeutics were involved in trial design, execution, and data analysis. Cidara Therapeutics and Mundipharma were involved in trial reporting.

Acknowledgements

Medical writing services were provided by Rebecca Down at Copperfox Communications Ltd.